A Phase 1b Study of IMU-131 HER2/neu Peptide Vaccine plus Chemotherapy in Patients with HER2/neu Overexpressing Metastatic or Advanced Adenocarcinoma of the Stomach or Gastroesophageal Junction

#4030

MEDICAL UNIVERSITY OF VIENNA

IMUGENE

Yee Chao[1], Thomas Cheung Yau[2], Marina Maglakelidze[3], Iurie Bulat[4], Suebpong Tanasanvimon[5], Chaiyut Charoentum[6], Wichit Arpornwirat[7], Jedzada Maneechavakajorn[8], Arunee Dechaphunkul[9], Teerapat Ungtrakul[10], Chia-Jui Yen[11], Li-Yuan Bai[12], Wen-Chi Chou[13], Ursula Wiedermann[14], Erika Garner-Spitzer[14] Anthony J Good[1516], Nick Ede[15], Leslie Chong[15]

1. Department of Oncology, Taipei Veterans General Hospital, Taipei, Taiwan 2. Department of Medicine, The University of Hong Kong, Queen Mary Hospital, Hong Kong, Hong Kong 3. ARENSIA Exploratory Medicine LLC, Tbilisi, Georgia 4. ARENSIA Exploratory Medicine Research Unit, Institute of Oncology, Chisinau, The Republic of Moldova 5. King Chulalongkorn Memorial Hospital, Bangkok, Thailand 6. Maharaj Nakorn Chiang Mai Hospital, Mueang Chiang Mai District, Thailand 7. National Cancer Institute, Bangkok, Thailand 8. Rajavithi Hospital, Bangkok, Thailand 9. University of Alberta, Edmonton, AB, Canada 10. Faculty of Medicine and Public Health, HRH Princess Chulabhorn College of Medical Science, Bangkok, Thailand 11. National Cheng Kung University Hospital, Tainan, Taiwan 12. China Medical University Hospital, Taichung, Taiwan 13. Linkou Chang Gung Memorial Hospital, Taoyuan, Taiwan 14. Medical University Vienna, Vienna, Austria 15. Imugene, Sydney, Australia 16. For correspondence: anthony.good@imugene.com

Abstract

Background: Gastric cancer is the 5th most common cancer and the 3rd leading cause of cancer deaths. HER2/neu is overexpressed in 15% - 25% of patients with gastric cancer. Monoclonal antibodies against HER2/neu are effective but alternatives are needed due to cost and global availability. IMU-131 is a B-cell peptide vaccine composed of a fusion of 3 epitopes from the extracellular domain of HER2/neu conjugated to CRM197 with the adjuvant Montanide. Polyclonal antibodies against IMU-131 peptides elicit antitumor activity in vitro and a phase I study demonstrated safety and immunogenicity in Her-2 +/++ breast cancer patients.

Methods: IMU-131 was given to patients with HER2/neu overexpressing gastric or gastroesophageal junction (GEJ) adenocarcinoma in an international open-label Phase 1b dose escalation trial performed in 14 Asian and Eastern European sites assessing safety, tolerability, and immunogenicity. Each patient received IMU-131 on Days 0, 14, and 35, accompanied by cisplatin and 5-fluorouracil or capecitabine every 21 days.

Results: 14 patients were enrolled with advanced stage IIIb or IV with 10 HER2 overexpressing tumors (7 x HER2+++, 3 x HER2++ FISH positive) and 4 HER2++ expressing tumors. Mean age was 57 yo (range of 21 - 79) with ECOG scores of 0 or 1 in 7 patients each. There were 9 Asian and 5 Caucasian patients with 5 females and 9 males. Dose levels were 10, 30 and 50 μg with 3, 6, and 5 patients receiving those dose levels each. 11 patients received all 3 doses with 3 patients who received only 2 doses due to disease progression and 2 patients received a dose on day 182.

Of the 14 patients dosed 11 were evaluable for tumor progression at day 56 and later. Of those patients, the best response was 1 CR, 4 PR,5 SD and 1 PD. In the 10 μg dose group the best response was 1 CR and 2 SD, with 2 PR, 2 SD and 1 PD in the 30 μg group and 2 PR and 1 SD in the 50 μg group. In patients with HER2 overexpression there was 1 CR, 4 PR, 2 SD and 1 PD, and in patients with HER2++ expression there was 3 SD. There were no SAEs related to IMU-131 and 1 patient had a mild injection site reaction.

Conclusions: IMU-131 is a promising B-Cell vaccine against HER2. Further work in a controlled phase 2 trial is ongoing.

Background

IMU-131 contains a single peptide antigen composed of 3 individual linear B-cell epitope peptide sequences selected from HER-2/neu (P4/P6/P7) that induce the patient's own B-cells to produce anti-HER-2/neu antibodies[4]. HER-2/neu is a tumor-associated protein that is overexpressed in several malignancies and has been proposed as a human cancer vaccine target[2]. Overexpression of HER-2/neu in human breast and gastric carcinomas correlates with a more aggressive course of disease[1] with poorer overall survival than in patients without overexpression of HER-2/neu[3]. IMU-131 may complement or replace the high-dose passive immunization induced by trastuzumab with a persistent humoral response to HER-2/neu. The immunological proof of principle has been demonstrated in mice by administering the 3 HER-2/neu epitopes as either individual or collinear peptides conjugated to different carriers and formulated with or without adjuvants. Vaccination of mice with 3 HER-2-peptides representing B-cell epitopes of the extracellular domain (ECD) of HER-2/neu induces HER-2/neu specific immunoglobulin G (IgG) antibodies with strong antitumor activity in vitro and in vivo[4][5].

Study Design

Patients had stage III or IV gastric or GEJ adenocarcinoma not amenable to surgical resection and had HER2/neu overexpression (3+ by IHC or IHC 2+ confirmed by FISH or CISH). The doses tested were 10, 30 and 50 μg (peptide P467 antigen equivalent) with injection volumes of 0.1, 0.3 and 0.5 mL on days 0, 14, and 35, with chemotherapy cycles every 21 days starting 14 days ($\pm$1 day) after first IMU-131 vaccination. Chemotherapy included Cisplatin, IV (80 mg/m^2 on Day 14, then every 21 days and either 5-FU, 4000 mg/m^2 CIV (1000 mg/m^2/day infusion for 96 hrs on days 14-17, then every 21 days) or Capecitabine for 14 days at 2000 mg/m^2/day,

Study Design cont.

orally (1000 mg/m^2 twice daily on days 14-27, then every 21 days). Patient's cellular (T_{reg}/T_{eff} and B_{reg}/B_{eff} cells), and humoral immunity (anti-P467 and anti-HER2 antibodies) were evaluated. Radiographic assessment evaluated disease progression (RECIST 1.1). Patients who completed Day 56 continued in long term maintenance with administration of IMU-131 on Day 98 then every 84 days.

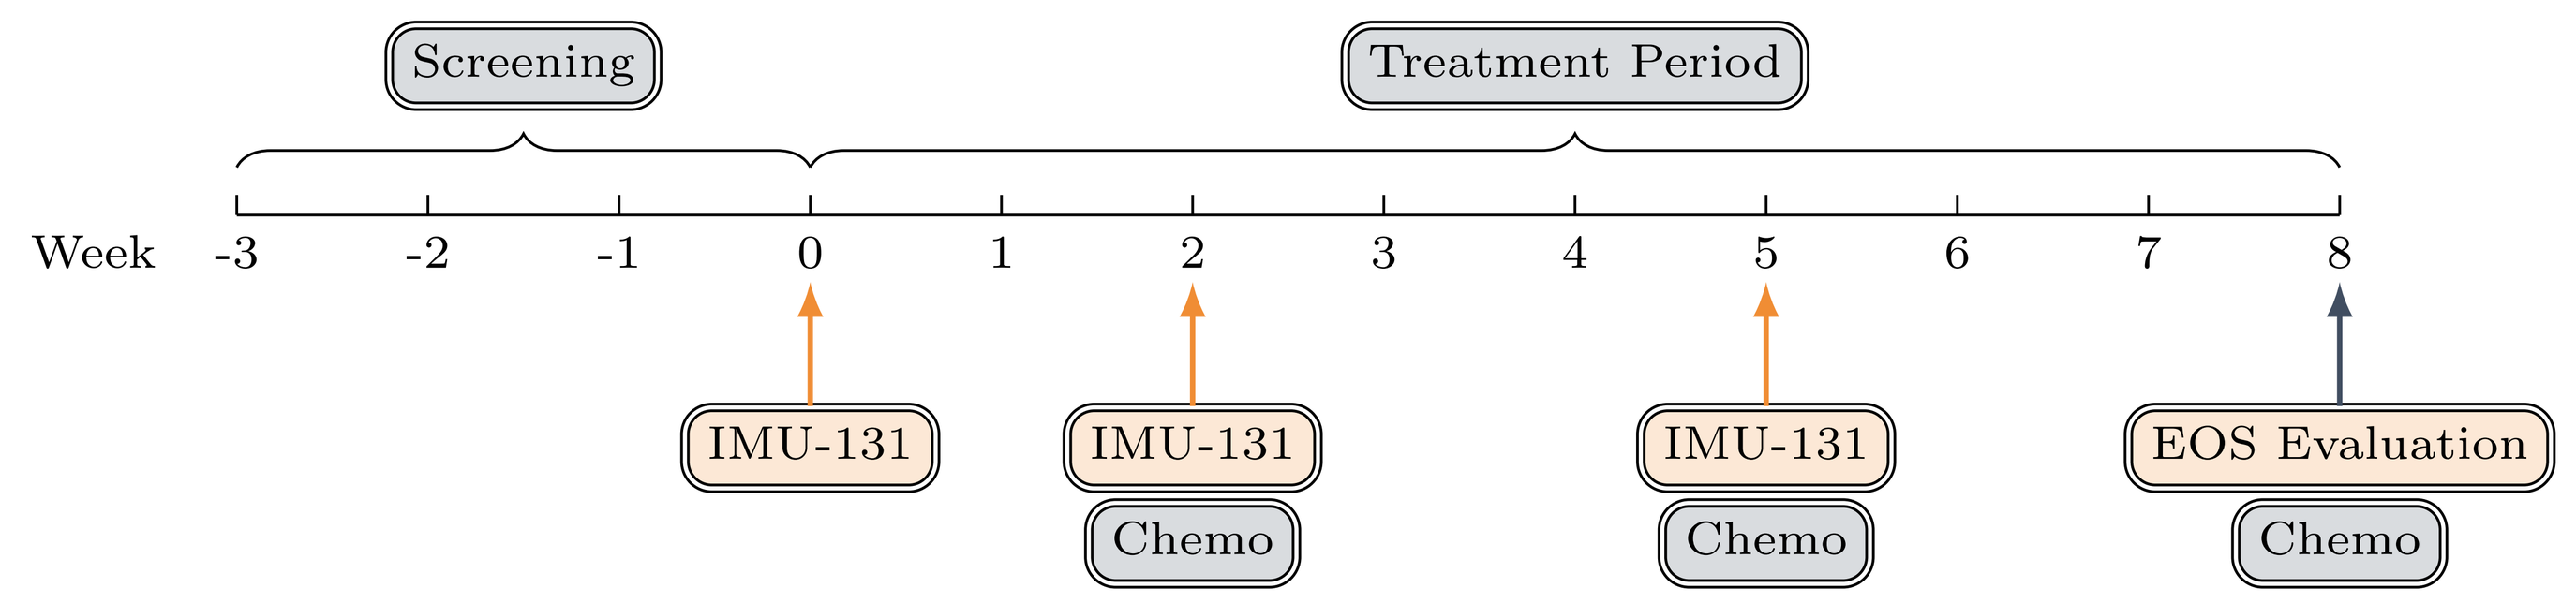

Screening
Treatment Period
Week -3 -2 -1 0 1 2 3 4 5 6 7 8
IMU-131
IMU-131
Chemo
IMU-131
Chemo
EOS Evaluation
Chemo

Results

Fourteen patients were enrolled from 14 sites in 5 countries (Georgia, Hong Kong, Republic of Moldova, Taiwan and Thailand). Three patients were enrolled in the 10 μg, 6 in the 30 μg and 5 in the 50 μg cohorts. Nine patients (64.3%) were Asian and 5 (35.7%) were Caucasian. All 14 patients were included in the safety population, 11 (78.6%) patients completed the treatment period, 3 (21.4%) patients discontinued treatment and 10 (71.4%) patients discontinued the study. At interim analysis, 1 patient in IMI-131 30 μg cohort and 3 patients in the IMI-131 50 μg cohort were continuing participation in the study. Five patients were (35.7%) female and 9 (64.3%) male with a mean age of 57 $\pm$ 17 yrs. Tumor stages were IIIb (n=2), IIIc (n=1) and IV (n=11).

Immunogenicity: Antibodies that cross-reacted with HER2/neu extracellular protein and the p467 protein construct were measured. For both antibodies there were dose and time dependent increases. Antibody levels tended to increase with each dose administration and were at their highest levels at Day 56.

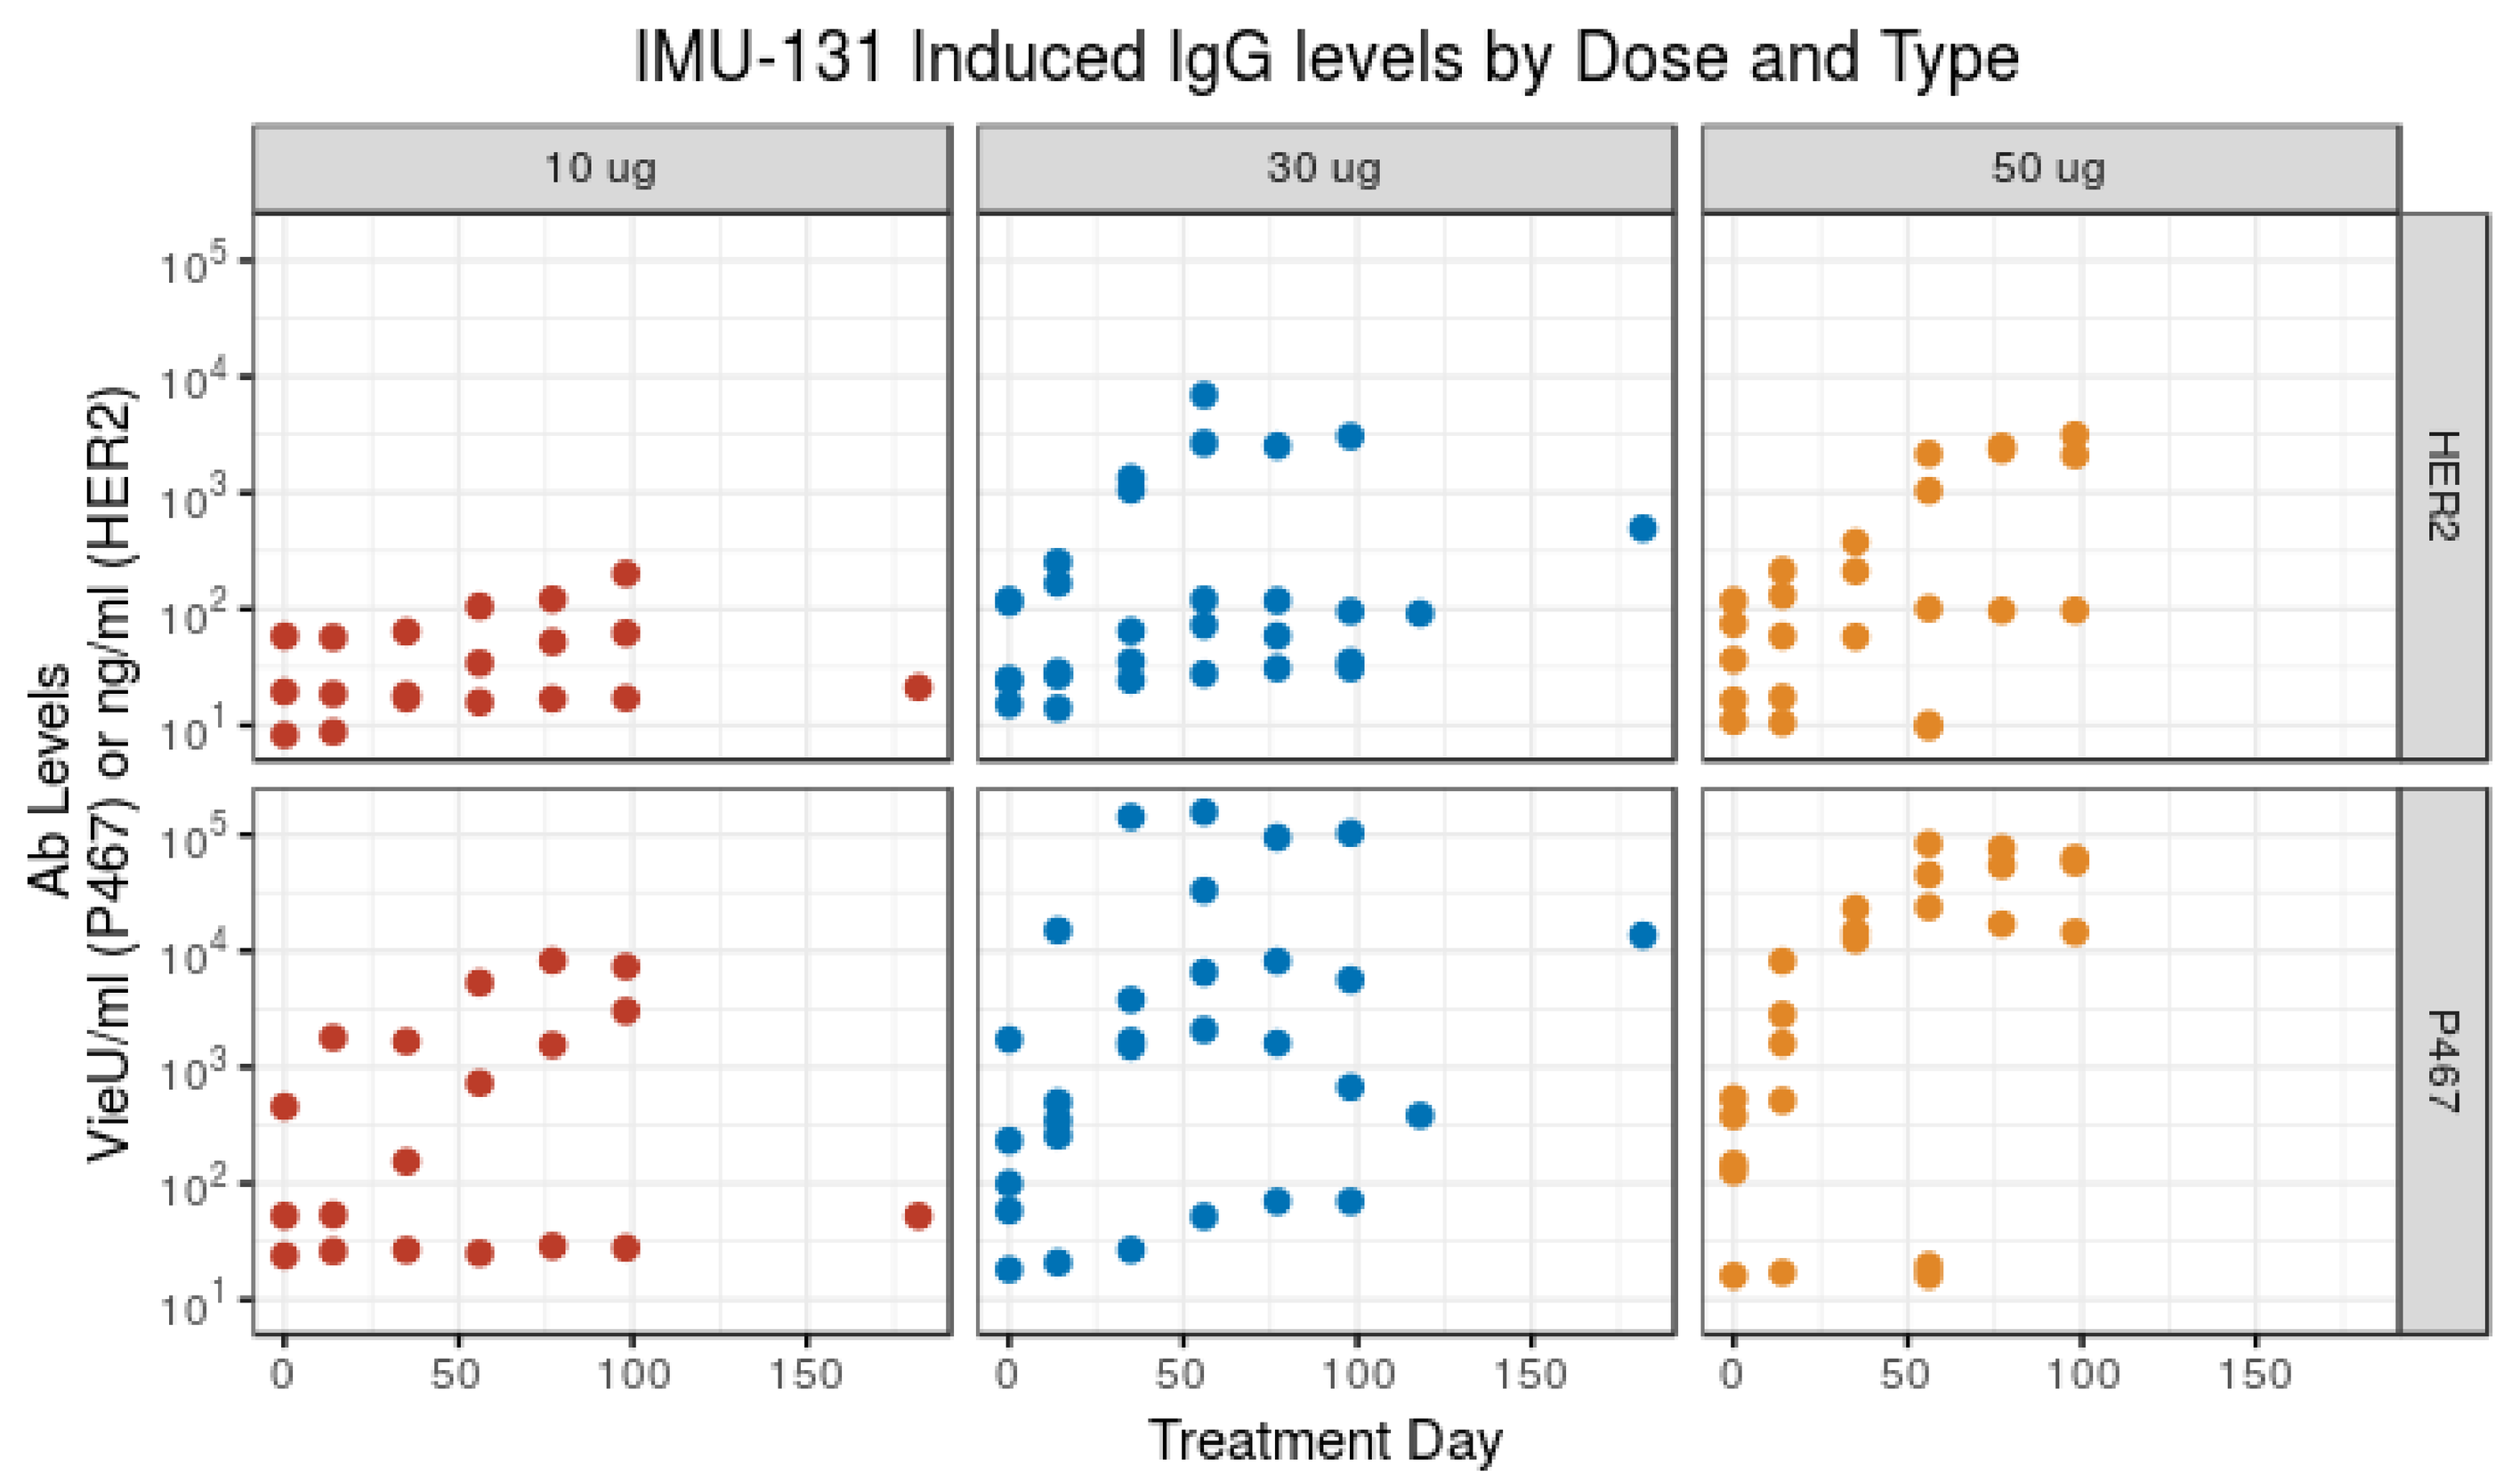

IMU-131 Induced IgG levels by Dose and Type
10 ug
30 ug
50 ug
HER2
P467
Ab Levels
VieU/ml (P467) or ng/ml (HER2)
Treatment Day

Tumor Response: Overall tumor size decreased from baseline over time. The overall mean percentage change at Visit Day 56 was -23.17% (SD = 27.10%), -25.10% (SD = 35.37%) at the start of long term maintenance Day 98 and -58.98% (SD = 6.28%) at long term maintenance Day 182. Tumor size decreases from baseline over time was also noted for all treatment groups.

Safety: Overall, 207 TEAEs were reported by 14 patients with majority of the events assessed as Grade 1 to 3 severity and not related to IMU-131. Two Grade 1 vaccination site reactions (injection site reactions) (pruritus and erythema) were reported by 1 patient in the IMU-131 50 μg treatment group which were assessed as possibly related to IMU-131 and

Results cont.

1 patient in the 30 μg reported 2 TEAEs (hypoalbuminemia and hyponatremia) that were assessed as related to IMU-131 and 1 patient in the IMU-131 30 μg reported 2 TEAEs (decreased appetite and weight decreased) that were assessed as possibly related to IMU-131. The TEAEs observed in this study were in line with and expected for the chemotherapy the patient received. A total of 9 patients reported 15 SAEs of which 5 TEAEs resulted in fatal outcomes. The TEAEs (convulsion, pneumonia, acute renal failure, dyspnea and acute respiratory failure) leading to fatal outcomes were not related to the study drug (IMU-131). No DLTs were observed and there were no events leading to study drug discontinuation. Clinical laboratory findings including vital signs, ECG and physical examinations were unremarkable and did not impact the overall safety results.

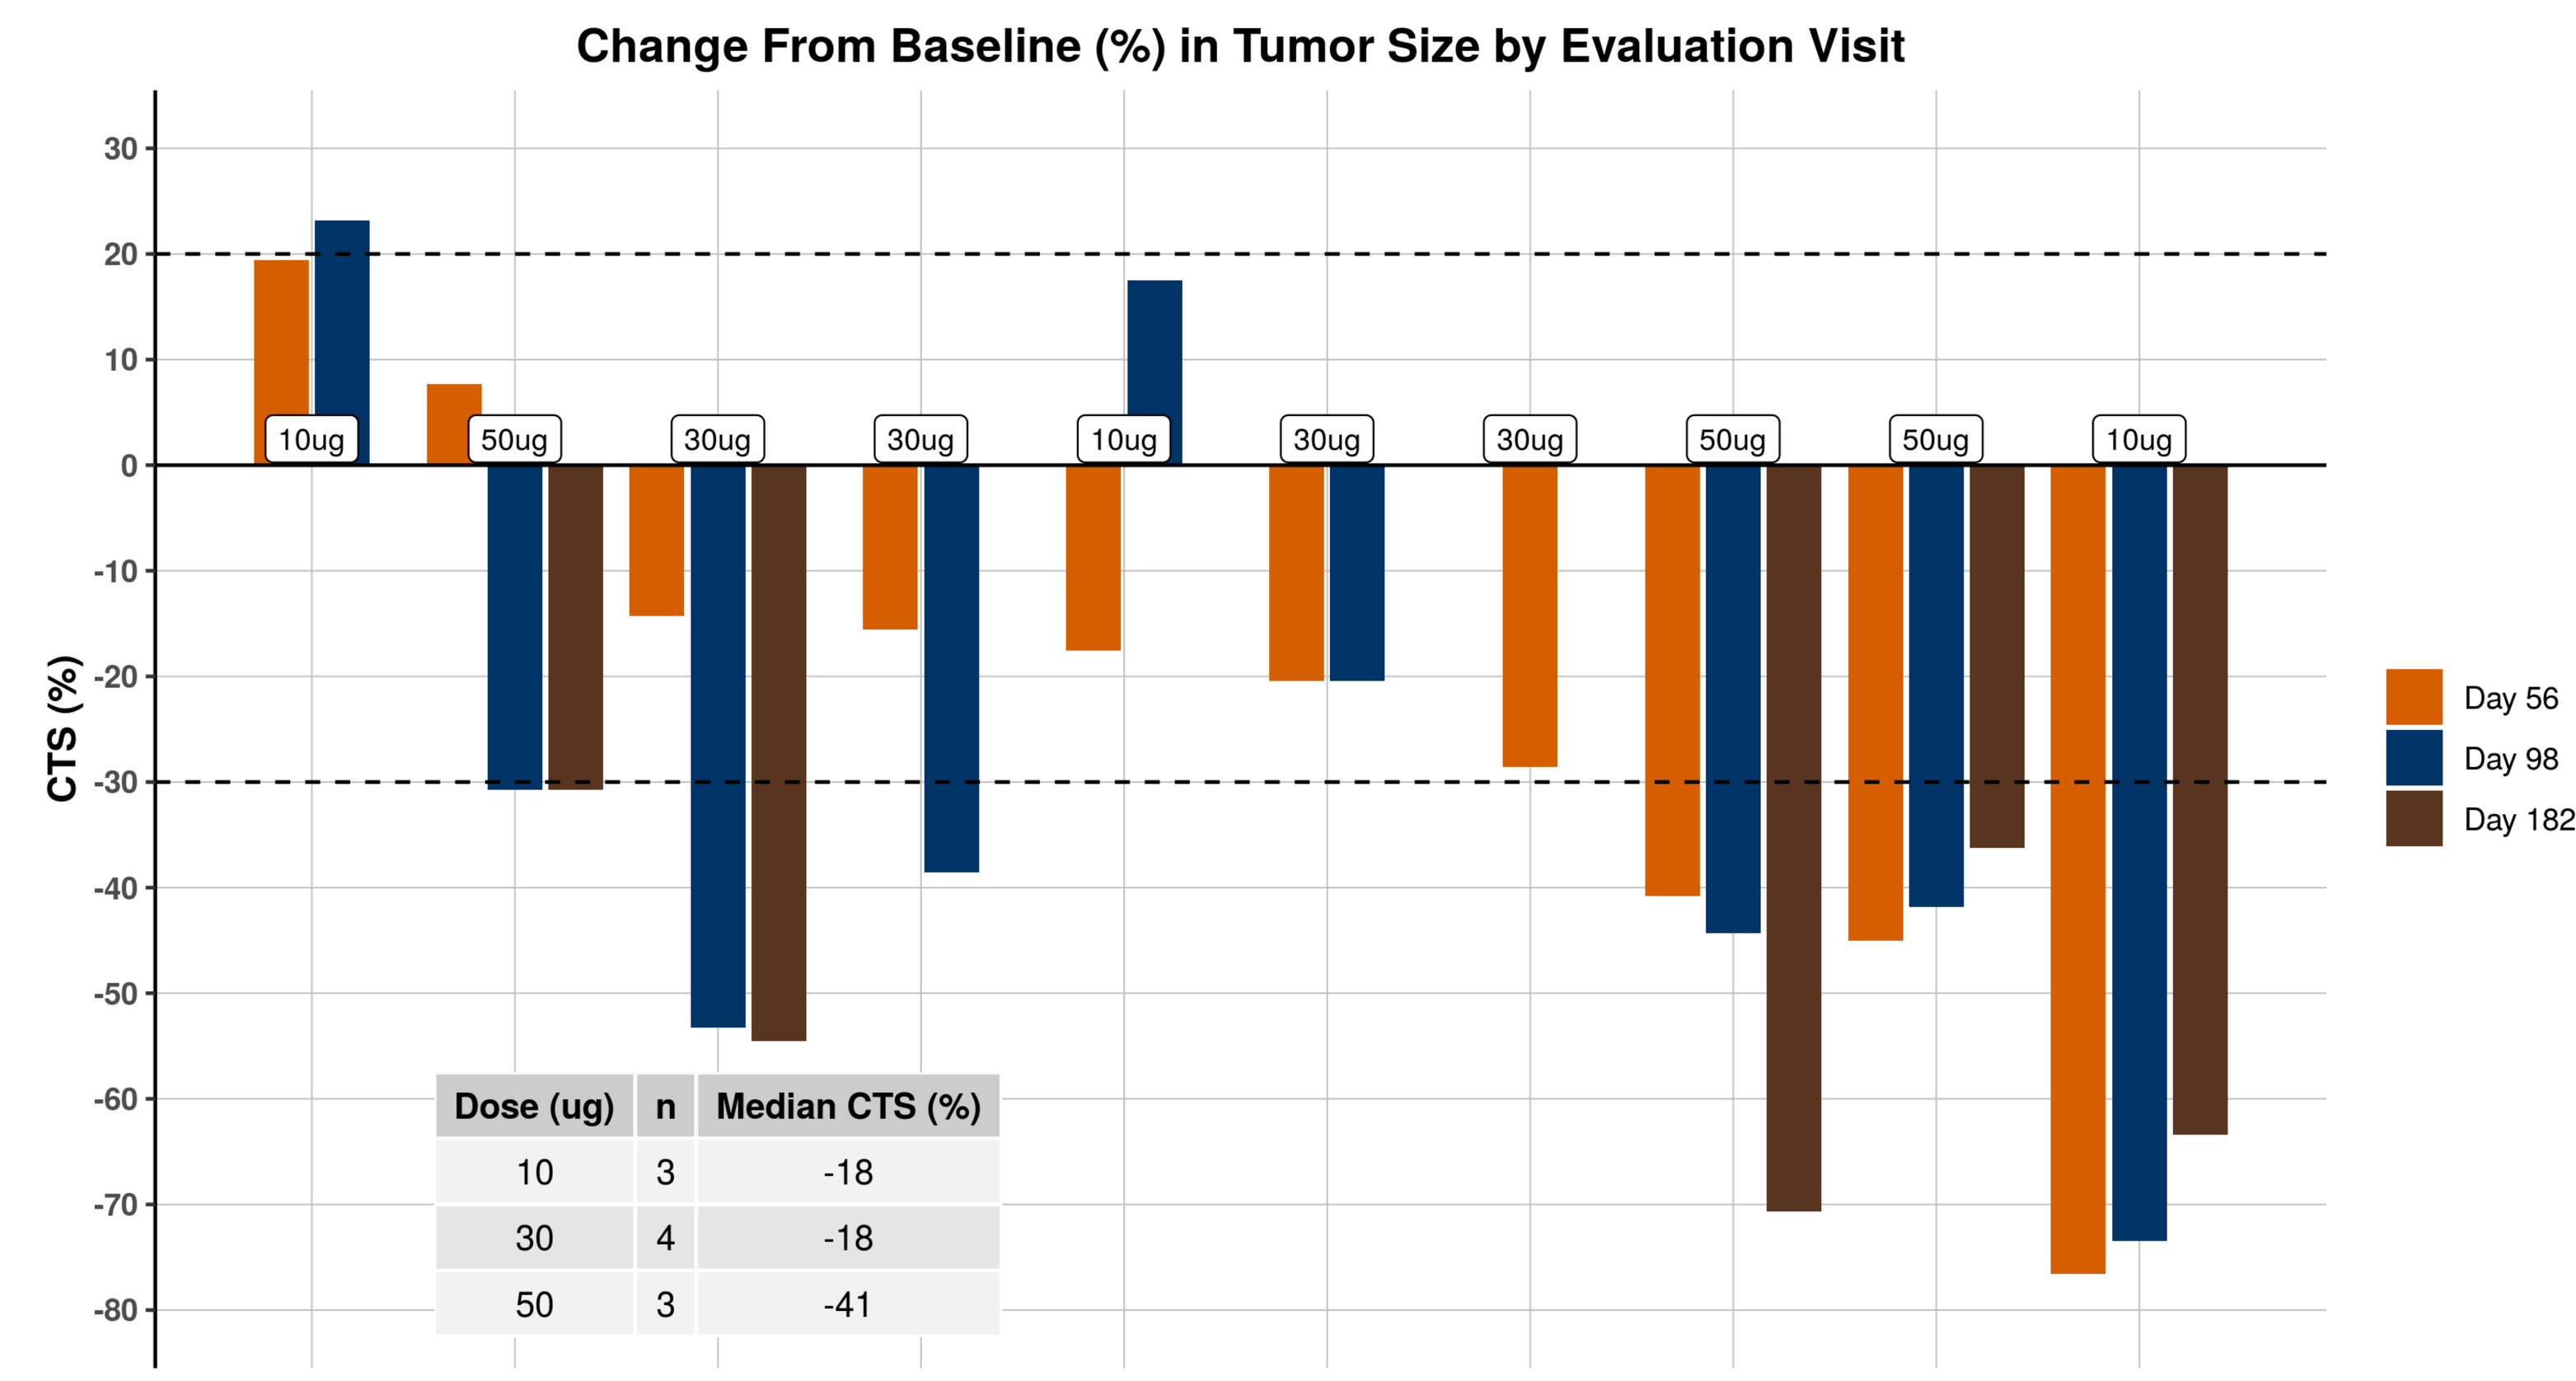

Change From Baseline (%) in Tumor Size by Evaluation Visit
CTS (%)
10ug
50ug
30ug
30ug
10ug
30ug
30ug
50ug
50ug
10ug
Day 56
Day 98
Day 182
Dose (ug) n Median CTS (%)
10 3 -18
30 4 -18
50 3 -41

Patient Tumor and Response Data

Patient	Dose (μg)	HER2 Status	Tumor Size[a] (RECIST) Baseline	Day 56	Day 98	Day 182
TH02 001	10	2+/FISH-	97	80 (SD)	114 (PD)	Withdrawn
TH03 001	10	2+/FISH-	268	320 (SD)	330 (PD)	Deceased
TW02 002	10	3+	38	9 (CR)	10 (PR)	14 (PD)
TH05 013	30	2+/FISH+	49	39 (SD)	39 (SD)	Deceased
TH05 015	30	3+	78	66 (SD)	36 (PR)	35 (PR)
GE01 004	30	3+	122	103 (SD)	75 (PR)	Deceased
TW01 001	30	3+	56	40 (PD)[b]	Withdrawn	Withdrawn
MD01 003	30	3+	NTL[c]	NTL (SD)	NTL (PD)	Withdrawn
TW02 003	50	2+/FISH+	66	36 (PR)	39 (PR)	42 (PR)[d]
MD01 007	50	2+/FISH -	26	28 (SD)	18 (PR)	18 (PR)[d]
TW02 004	50	3+	177	105 (PR)	99 (PR)	52 (PR)[d]

[a] Sum of the diameters of the target lesions [b] Due to 2 new lesions [c] Non-target lesions
[d] Ongoing in Study

Conclusion

The preliminary immunology and clinical response data are promising. Safety data indicate that IMU-131 is well-tolerated with no significant local or systemic reactions. There were no dose-limiting toxicities observed, no significant injection site reactions and no IMU-131 related SAEs. The 50 μg dose produced the most consistent P467 and HER-2 specific antibodies compared to 10 and 30 μg doses with preliminary response data demonstrating 50 μg of IMU-131 was associated with tumor size reduction. The 50 μg dose of IMU-131 will be used in a phase 2 study.

References

[1] In: *Oncogene* 9 (8 Aug. 1994).[2] In: *Journal of immunology* 156 (9 May 1996).[3] In: *Microsc Res Tech* 59 (2 Oct. 2002).[4] In: *Int J Cancer* 107 (6 Dec. 2003).[5] In: *Breast Cancer Res Treat* 106 (1 Nov. 2007).