Abstract CT 017: Phase Ib Immunotherapy Trial with a Combination of Two Chimeric (Trastuzumab-like and Pertuzumab-like) HER-2 B Cell Peptide Vaccine emulsified in ISA 720 and nor-MDP Adjuvant in Patients with Advanced Solid Tumors, Immunological Response and Clinical Outcome

Bekaii-Saab T[1], Wesolowski R[2,6], Ahn DH[1], Wu C[3], Mortazvi A[2,6], Lustberg M[2,6], Fowler J[4], Wei L[5,6], Overholser J[4], Kaumaya PTP[4,6]

[1]Division of Hematology and Medical Oncology, Mayo Clinic, Phoenix AZ; [2]Division of Medical Oncology, Ohio State University, Columbus OH; [3]Department of Hematology & Medical Oncology, Emory University School of Medicine, Atlanta GA; [4]Department of Obstetrics & Gynecology, Columbus OH; [5]Center for Biostatistics, [6]Arthur G. James Cancer Hospital/Comprehensive Cancer Center, Columbus, Ohio. Columbus OH

Abstract

While combined anti HER-2 directed therapy is active and well-tolerated across solid tumor malignancies, studies investigating novel agents and combination therapies with anti HER-2 directed therapy, are also under investigation. One strategy of interest has been investigating the potential therapeutic role of HER-2 peptide cancer vaccines, which has the potential to result in a sustained humoral immunity.

We have advanced a new paradigm in immunotherapy that focuses on humoral responses based on conformational B-cell epitope vaccines. These novel platforms elicit high affinity anti-peptide antibodies against tumors that help circumvent intrinsic drug resistance. Previously, we identified the first generation of HER-2 B-cell peptide epitopes and through computer immunogenicity algorithms and extensive *in vitro* and *in vivo* preclinical studies. In a recently published phase 1 clinical trial, we showed that the combination of the two chimeric HER-2 vaccine in patients with metastatic solid tumors was safe, demonstrated activity (disease control rate of 24%), and elicited HER-2 specific humoral responses in 62.5% of patients. Our group has developed two novel B-cell epitope specific vaccines consisting of epitopes derived from the extracellular domain of the HER-2/neu molecule that are binding sites of trastuzumab and pertuzumab.

In extensive preclinical studies, we have demonstrated that the two engineered epitopes mimicked the three-dimensional structure of the corresponding native HER-2 sequence and elicited high affinity antibodies that recognized the native HER-2 receptor. Additionally, these peptides inhibited multiple signaling pathways including HER-2 specific inhibition of cellular proliferation and cytoplasmic receptor domain phosphorylation. The peptide antibodies mediated antibody-dependent cellular cytotoxicity (ADCC). These vaccines had statistically reduced tumor onset in two transplantable tumor models (FVB/n and BALB/c) and led to significant reduction in tumor development in two transgenic mouse tumor models (BALB-*neu*T and VEGF$^{+/-}$Neu2–5$^{+/-}$). Lastly, these two vaccines exhibited properties similar to trastuzumab and pertuzumab, validating their use in this phase 1 clinical trial. Peptide vaccines offer an attractive immunotherapeutic option in the treatment of cancer, with considerable advantages in their safety, cost-effectiveness and ease of administration. Additional advantages of active cancer vaccines are exquisite specificity, low toxicity, and the potential for a durable treatment effect due to immunologic memory.

Herein, we report the results from the first-in-human, dose escalation portion of the phase Ib study testing the combination of two peptide B-cell epitope vaccine MVF-HER-2 (597-626) and MVF-HER-2 (266-296) incorporating a promiscuous measles virus (MVF) T cell epitope engineered to represent the trastuzumab and pertuzumab binding sites.

Objectives

- The primary objectives were to assess safety and clinical toxicity of immunization, determine the optimum immunologic /biologically dose (OID/OBD) of combination HER-2 vaccines,
- measure both humoral and cellular immune responses including the specificity, class and kinetics of anti-HER-2 peptide
- Secondary objectives were to collect and analyze post-immune sera and peripheral blood cells for additional 6 months following the last injection and document clinical responses
- Evaluate whether the combination of HER-2 vaccines demonstrate therapeutic benefit, provide synergistic and/or additive effects, and to enumerate mechanisms of action.

Study design and methods

- The GMP peptides were purchased from Peptisynthia (Torrance, CA) and acquired by Solway Group (Zug, Switzerland), and
- The GMP peptides met all the FDA and US Pharmacopeia requirements for sterility (ie, bacterial/fungal), endotoxins, and potency. Endotoxin levels of these peptides were tested and determined to be within acceptable levels as (GMP) grade.
- N-acetylglucosamine-3yl-acetyl-L-alanyl-D-isoglutamine (nor-MDP) was purchased from Penninsula Labs, (Torrance, CA).
- The vehicle Montanide ISA™ 720 was purchased from SEPPIC (Fairfield, NJ), and it had an approval certificate of analyses for toxicity, emulsifying property, and sterility
- The immunogenicity of each individual peptide and combination was verified in pairs of New Zealand rabbits.
- The combination vaccine was prepared by dissolving and mixing the two HER-2 B-cell epitope peptides with *n*-MDP adjuvant (0.025mg) in a total volume of 1.0 ml and emulsified 1:1 with ISA 720

The dose escalation part of this clinical trial used a 3+3 schema.
- 6 patients at each dose level were required to receive 3 inoculations of the combination vaccines at 3-week intervals.
- The dose levels (DLs) were 1.0 mg (DL1), 1.5 mg (DL2), 2.0 mg (DL3) and 2.5 mg (DL4) of each peptide.
- Initially, 3 patients were treated at each DL and observed for a minimum of 4 weeks. Dose escalation could proceed after 6 patients in a cohort were observed for a minimum of 4 weeks, and if only 0-1 of the 6 patients in that DL experienced DLT.

Immunization Schedule
- All eligible patients underwent a skin test prior to the first dose of the study vaccine. Approximately 0.1 mL of each peptide was injected intra dermally in two separate areas of the forearm. Patients were subsequently examined for delayed-type hypersensitivity (DTH) to the test dose approximately 20 minutes after administration (>20 mm erythema or development of a wheel).
- Patients who did not develop hypersensitivity reaction to the test dose, received 3 intramuscular vaccinations, each given 21 days apart (days 1, 22 and 43).
- The vaccine was injected into the gluteus maximus muscle, with subsequent injections given in contralateral muscle to the prior vaccine administration site.
- Following three doses, patients with responding or stable disease had an option of returning to receive booster vaccinations at 6 month intervals.

The clinical trial was conducted under an investigational new drug application (IND#14633,P.Kaumaya) approved by the FDA. The study protocol (2010C0075) was approved by the Ohio State University Institutional Review Board.
All patients gave written informed consent prior to participation in the study Clinical trials .gov (NCT 01376505).
The study was conducted in accordance with ethical principles founded in the Common Rule, the Belmont report, Good Clinical Practice guidelines, and applicable local laws.

Results

Fig. 1 Radiographic response

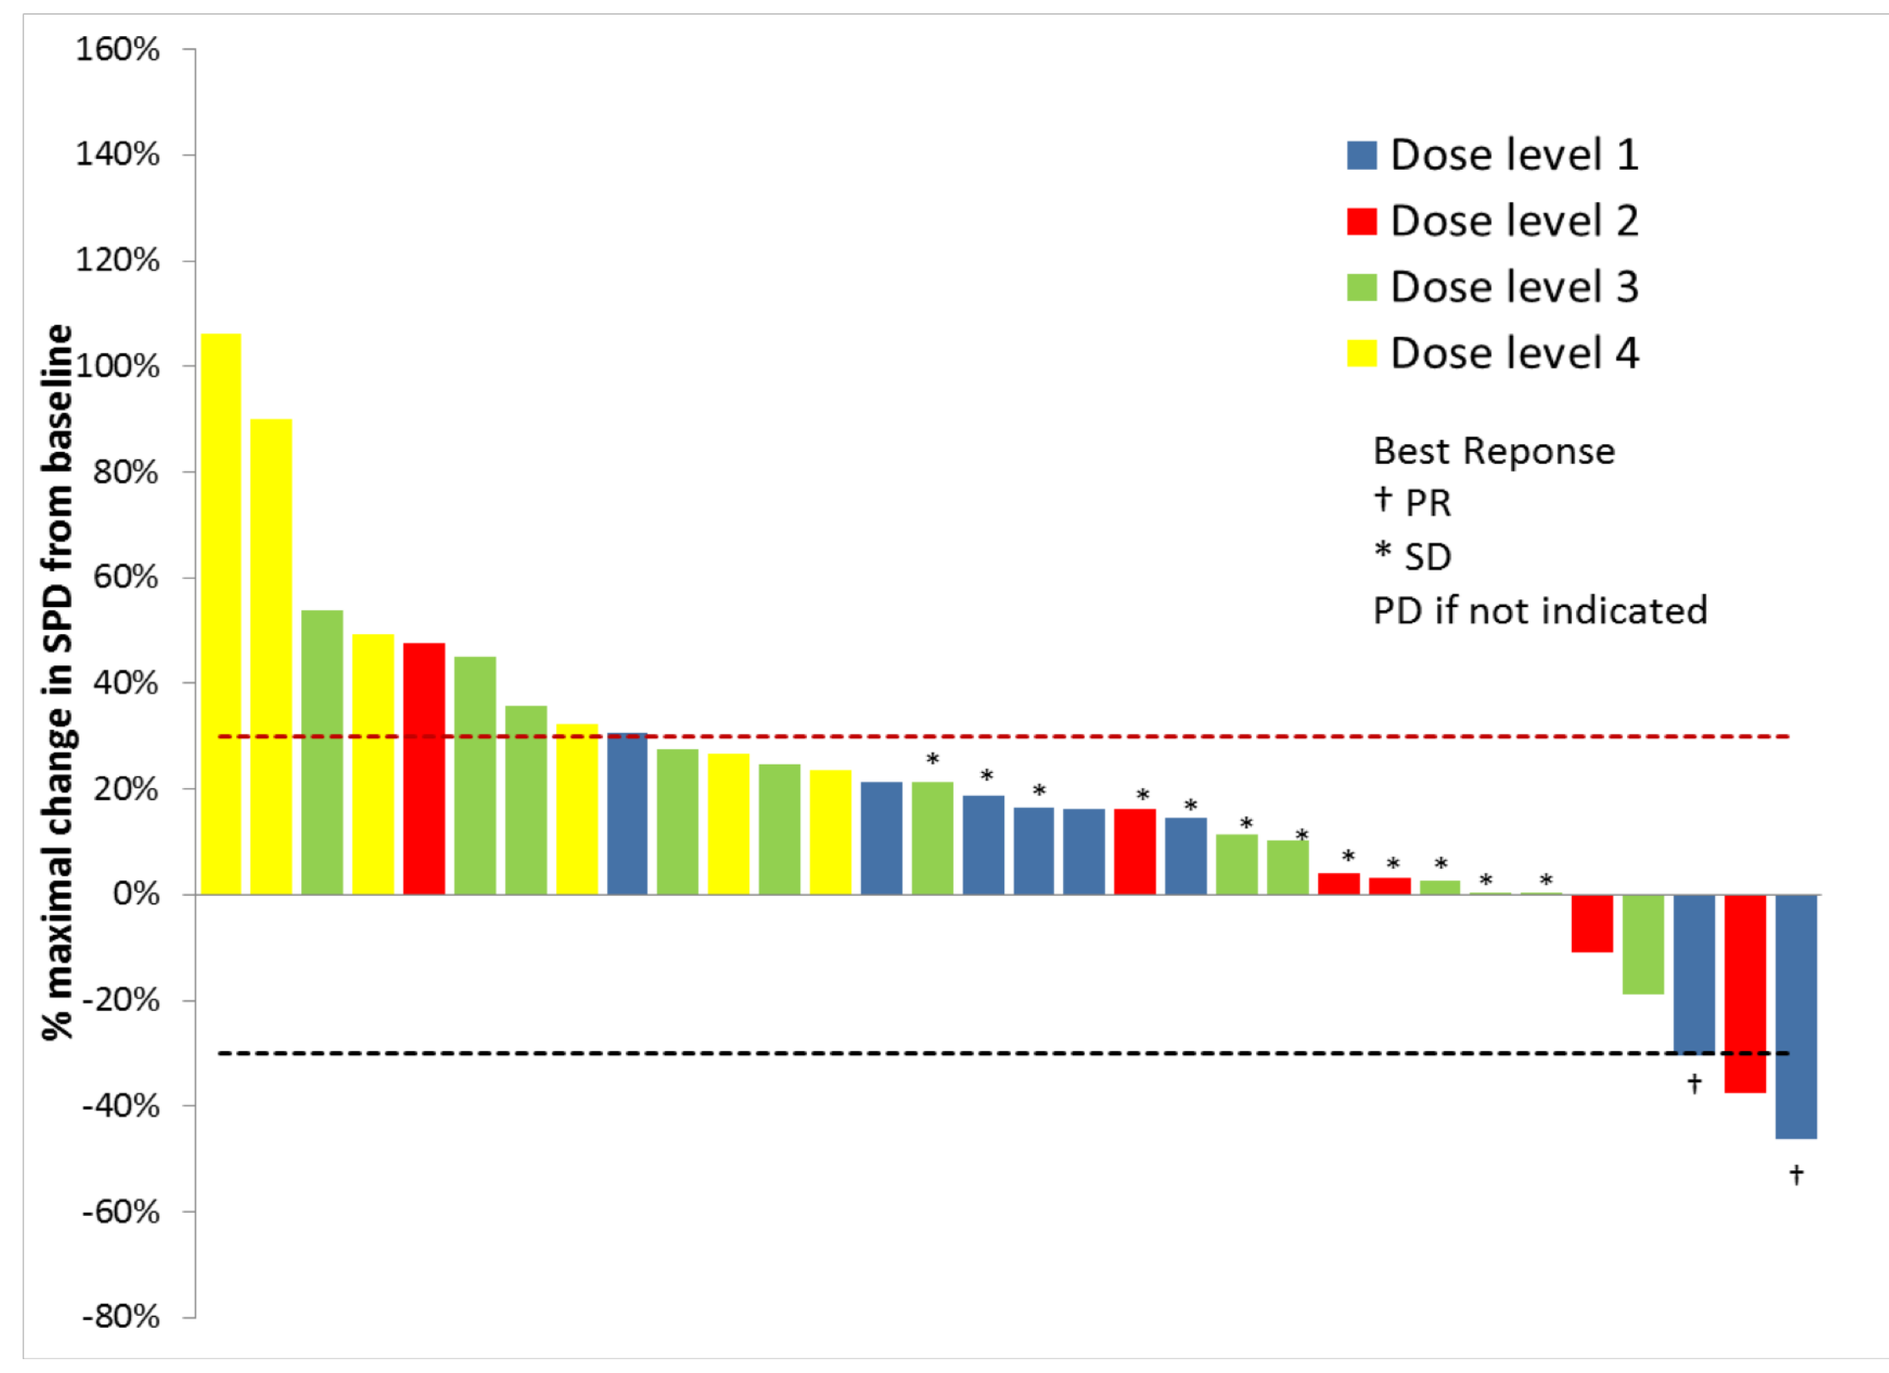
% maximal change in SPD from baseline
Dose level 1
Dose level 2
Dose level 3
Dose level 4
Best Reponse
† PR
* SD
PD if not indicated

Fig. 2 Duration of response

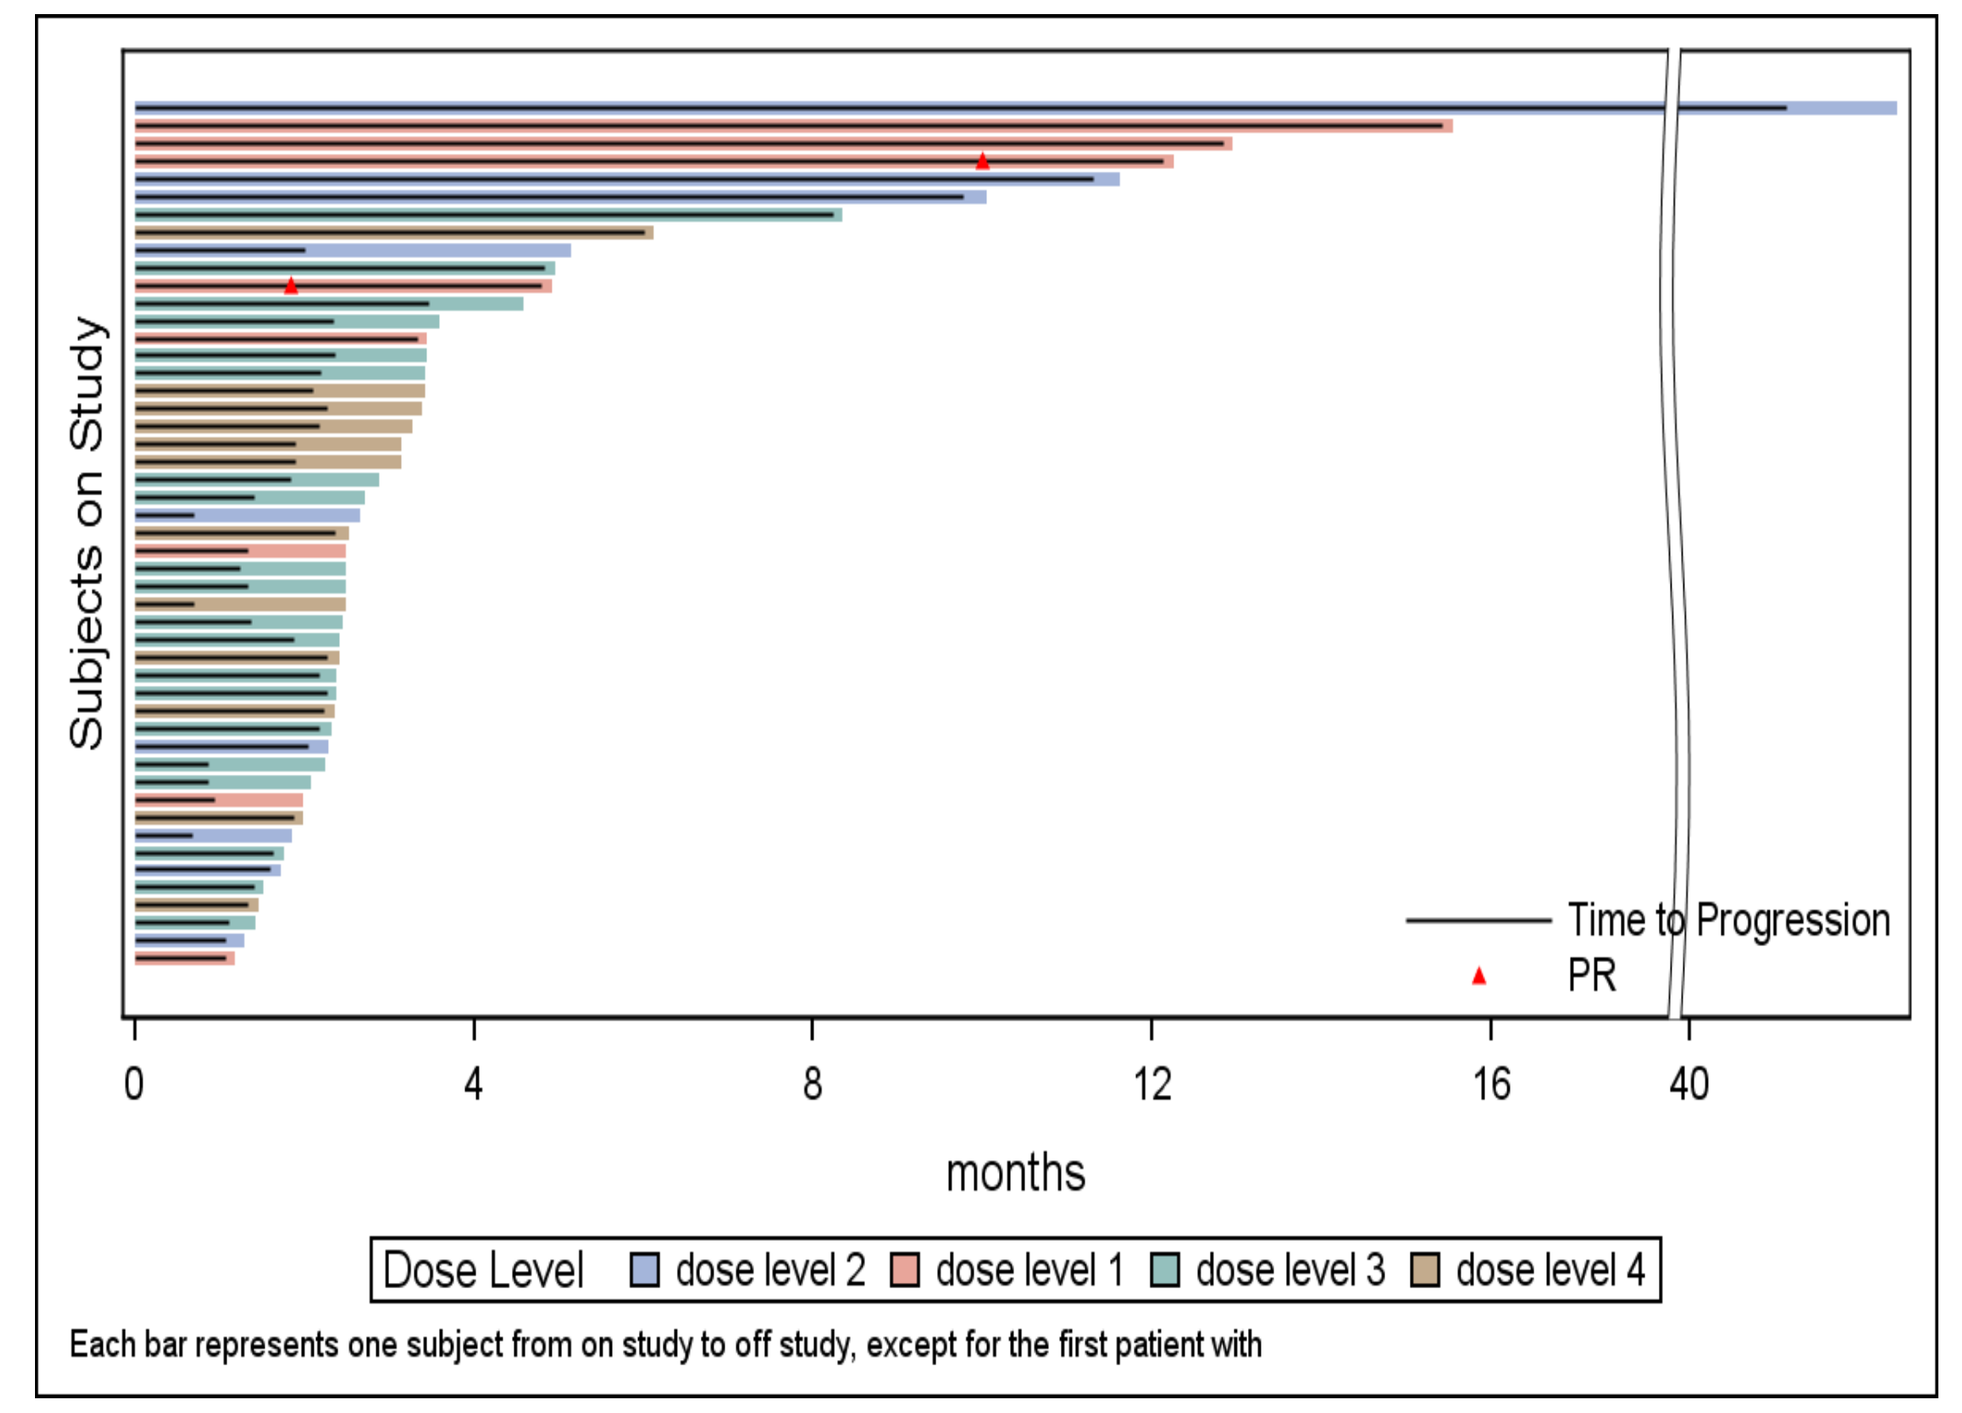
Subjects on Study
Time to Progression
PR
months
Dose Level
dose level 2
dose level 1
dose level 3
dose level 4
Each bar represents one subject from on study to off study, except for the first patient with

Fig 1 This waterfall plot illustrates the maximum percentage of tumor reduction for target lesions in the 31 patients with evaluable disease for tumor response. Each bar represents an individual patient. The red dash line represents the cut-off for disease progression, and the black dashed line represents the cut-off for partial response defined by RECIST 1.1.
Fig 2: This swimmers plot illustrates the survival of patients. Overall suvival (time to death) was determined from date of starting the study to the date of death. Time to progression is indicated for the patients with progression disease. Time to off study is also illustrated in the swimmer plot.

Fig. 3 Cohort 2 patients elicits native HER-2 IgG (BT474) and recombinant HER-2 anti-peptide antibodies

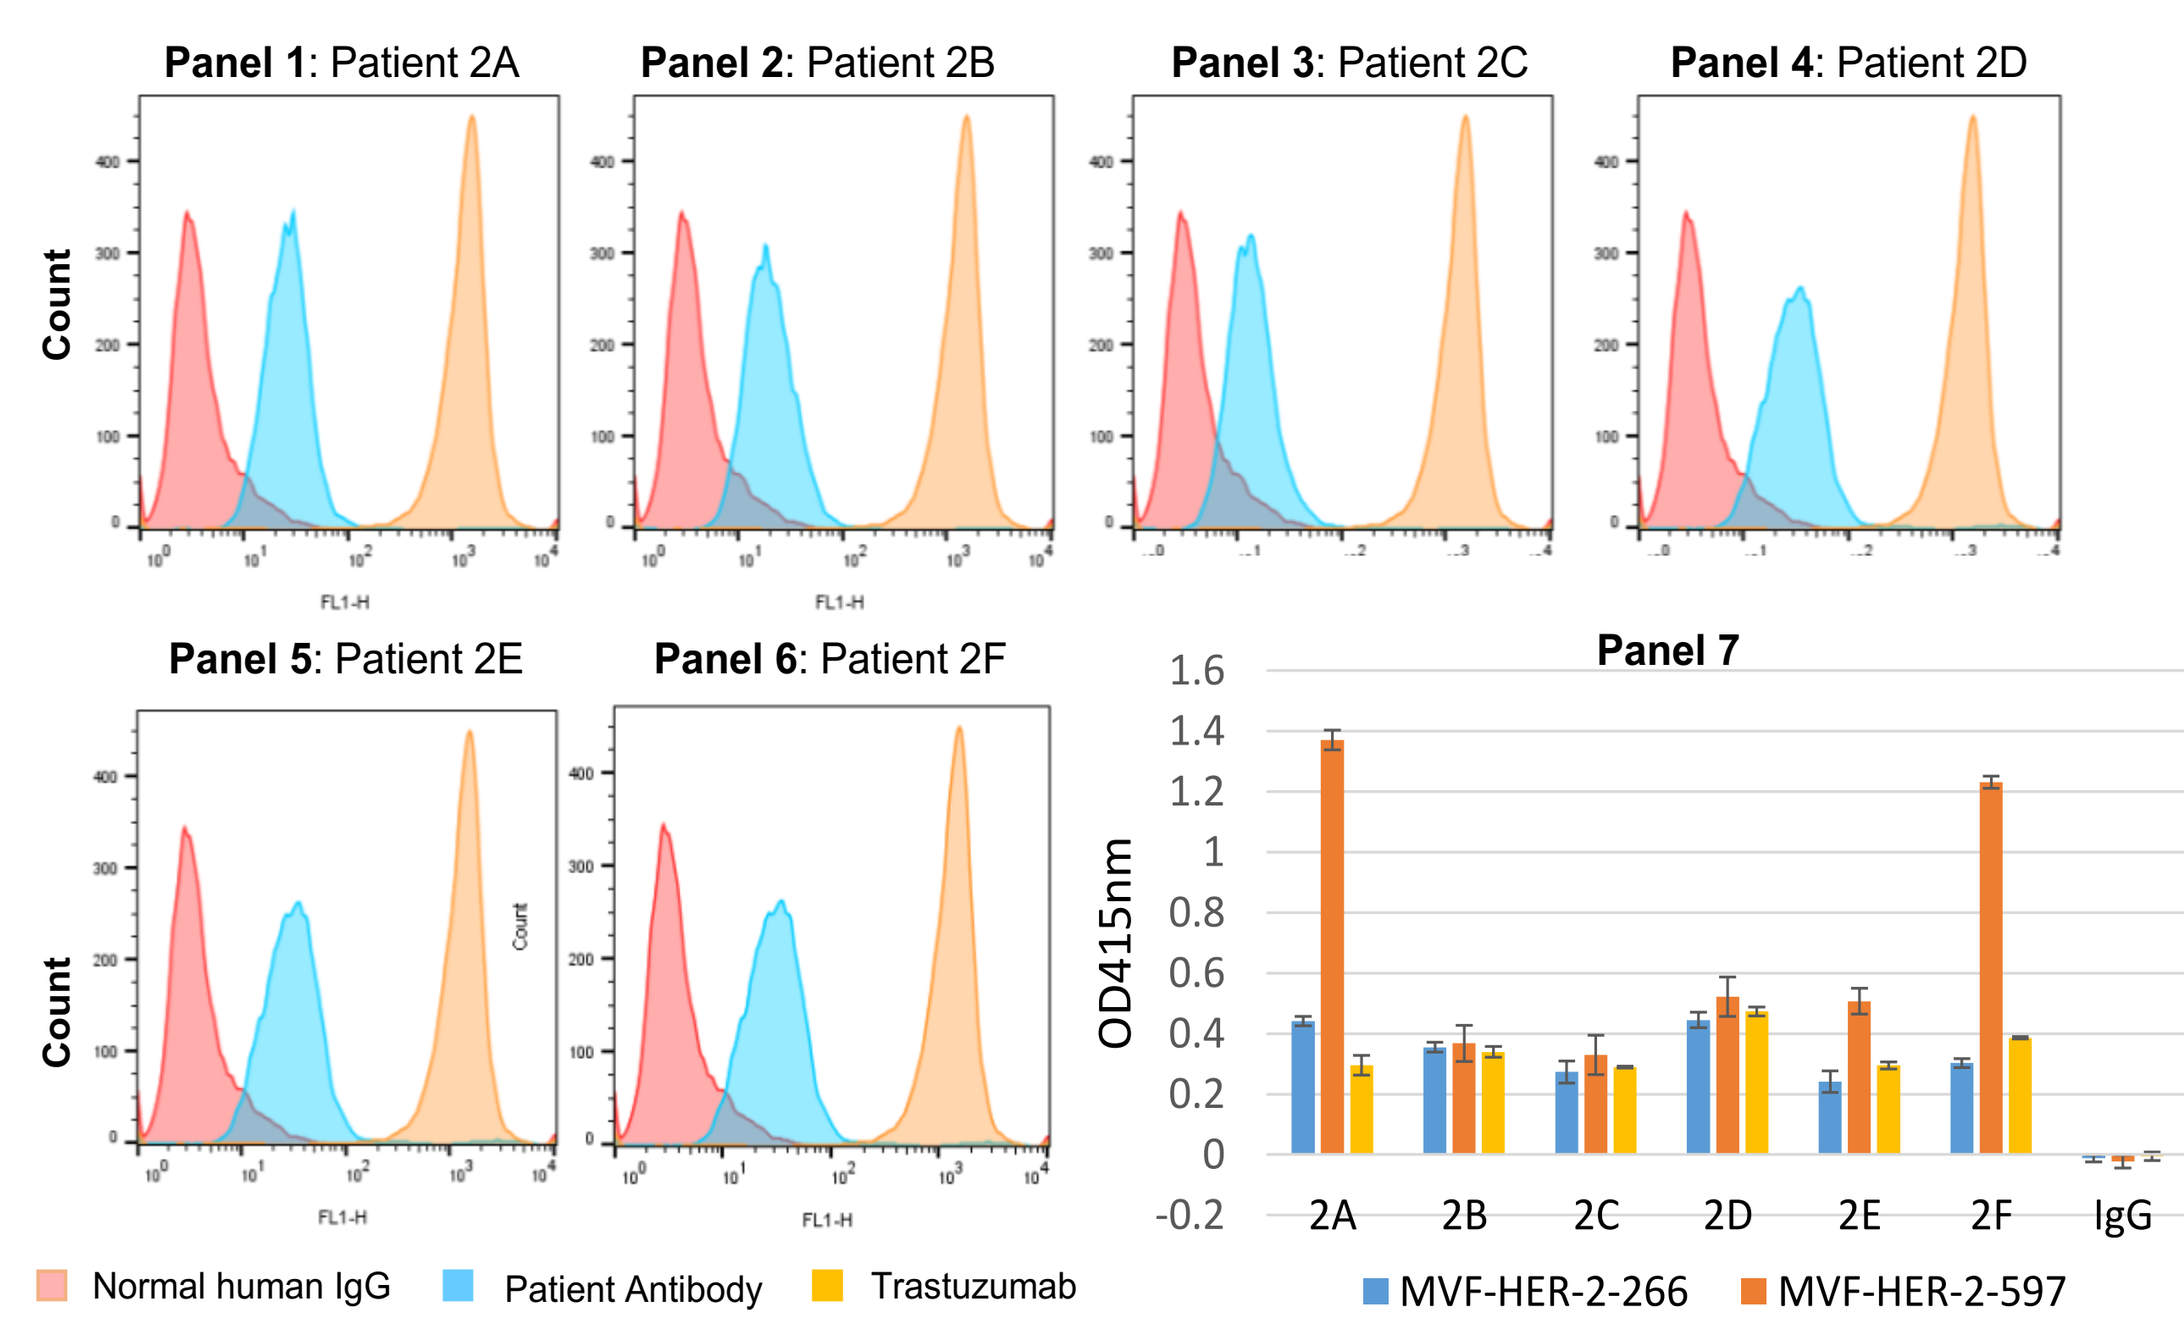
Panel 1: Patient 2A
Panel 2: Patient 2B
Panel 3: Patient 2C
Panel 4: Patient 2D
Panel 5: Patient 2E
Panel 6: Patient 2F
Panel 7
Count
OD415nm
Normal human IgG
Patient Antibody
Trastuzumab
MVF-HER-2-266
MVF-HER-2-597

Fig. 3 Panels (1-6) Flow cytometry analysis of cohort 2 patient (2A-2F) antibodies binding to BT-474 after the 3rd vaccination. 5x10 ^{5} BT-474 cells were incubated with 500µg/ml patient antibodies, 500µg/ml control antibody (normal human IgG) and 50µg/ml commercial human anti-HER2 (Trastuzumab) in staining buffer (PBS+0.05%BSA+0.2% NaN_3) respectively at 4°C for 2 hours and then with 1.6 µg/ml Alexa Fluor® 488 anti-human IgG at 4°C for 45min.
Panel 7 Binding of Cohort 2 antibodies to MVF-HER-2 (266-296) (blue), MVF-HER-2 (597-626) (orange) and recombinant HER-2 (yellow) assessed using ELISA. Antibodies purified via protein A/G column from sera before or after immunization were diluted to 20µg/ml with PBS/HS and incubated with immobilized MVF-HER-2 vaccines and recombinant HER-2. Bound antibodies were probed using HRP conjugated anti-human IgG. IgG, normal human IgG.

Table 1. Summary of patients in cohort 1 & 2

ID#	Cohort	# of Prior Therapies	Primary Cancer	Vaccinations (# of cycles + # of boosts)	HER-2/EGFR Status	Previous treatment type	Best Response
01-01	1	7	Colon	1	-ve/-ve	MC	PD
01-02		2	Anal	2	Unknown	MC	PD
01-03		1	Cartilage	3 +1	1A/ -ve/-ve	MC	SD
01-04		7	Lung	3 +1	1B/ -ve/-ve	MC	SD
01-05		1	Breast	3 +1	1C/ +ve/+ve	MC+ Trastuzumab	PR
01-06		3	Bladder	3	1D/Unknown	MC	PD
01-07		11	Ovarian	3	1E/ -ve/+ve	MC	PR
01-08		4	Colon	2	1F/ -ve/+ve	MC	SD
02-01	2	3	Anal	1	Unknown	MC	PD
02-02		5	Colon	2	-ve/+ve	MC	PD
02-03		3	Bladder	1	-ve/+ve	MC	SD
02-04		2	Anus	3 +1	2A/ -ve/+ve	MC	SD
02-05		3	Colon	2	2B/-ve/+ve	MC	PD
02-06		0	Parotid	3 +7	2C/ +ve	Radiation only	NE
02-07		3	Colon	3 +1	2D/ -ve/+ve	MC	SD
02-08		3	Breast	3	2E/ +ve/+ve	MC+ Trastuzumab	PD
02-09		8	Colon	3	2F/ Unknown	MC	SD

MC=multiple chemotherapy treatments; PR= partial response; PD=progressive disease; NE=non-evaluable; SD=stable disease; -ve=negative; +ve=positive. We have utilized standard RECIST 1.1 criteria to define PR, PD and SD
Cohorts 3 and cohorts 4 are not shown

Table 2. Treatment related toxicities

Toxicity (number of patients %	Grade 1-2	Grade 3-4	Total
Lymph node pain	1 (2%)	0 (0%)	1 (2%)
Fatigue	4 (8%)	0 (0%)	4 (8%)
Fever	2 (4%)	0 (0%)	2 (4%)
Flu like symptoms	1 (2%)	0 (0%)	1 (2%)
Injection site reaction	12 (24%)	0 (0%)	12 (24%)
Allergic reaction	1 (2%)	0 (0%)	1 (2%)
Alanine transaminase elevation	1 (2%)	0 (0%)	1 (2%)
Alkaline phasphagase elevation	1 (2%)	0 (0%)	1 (2%)
Lymphopenia	2 (4%)	0 (0%)	2 (4%)
Leukopenia	1 (2%)	0 (0%)	1 (2%)
Hypoalbminemia	2 (4%)	0 (0%)	2 (4%)
Hyponatremia	2 (4%)	0 (0%)	2 (4%)
Hypophosphatemia	0 (0%)	1 (2%)	1 (2%)
Buttock pain	1 (2%)	0 (0%)	1 (2%)
Myalgia	2 (4%)	0 (0%)	2 (4%)
Dry skin	1 (2%)	0 (0%)	1 (2%)
Pain of skin	1 (2%)	0 (0%)	1 (2%)
Pruritus	1 (2%)	0 (0%)	1 (2%)
Maculopapular Rash	1 (2%)	0 (0%)	1 (2%)
Skin and subcu. tissue disorder	3 (6%)	0 (0%)	3 (6%)
Skin ulceration	1 (2%)	0 (0%)	1 (2%)
Hematoma	1 (2%)	0 (0%)	1 (2%)

Fig. 4 Patient 2C antibody binding over 7 boosters (3.5years)

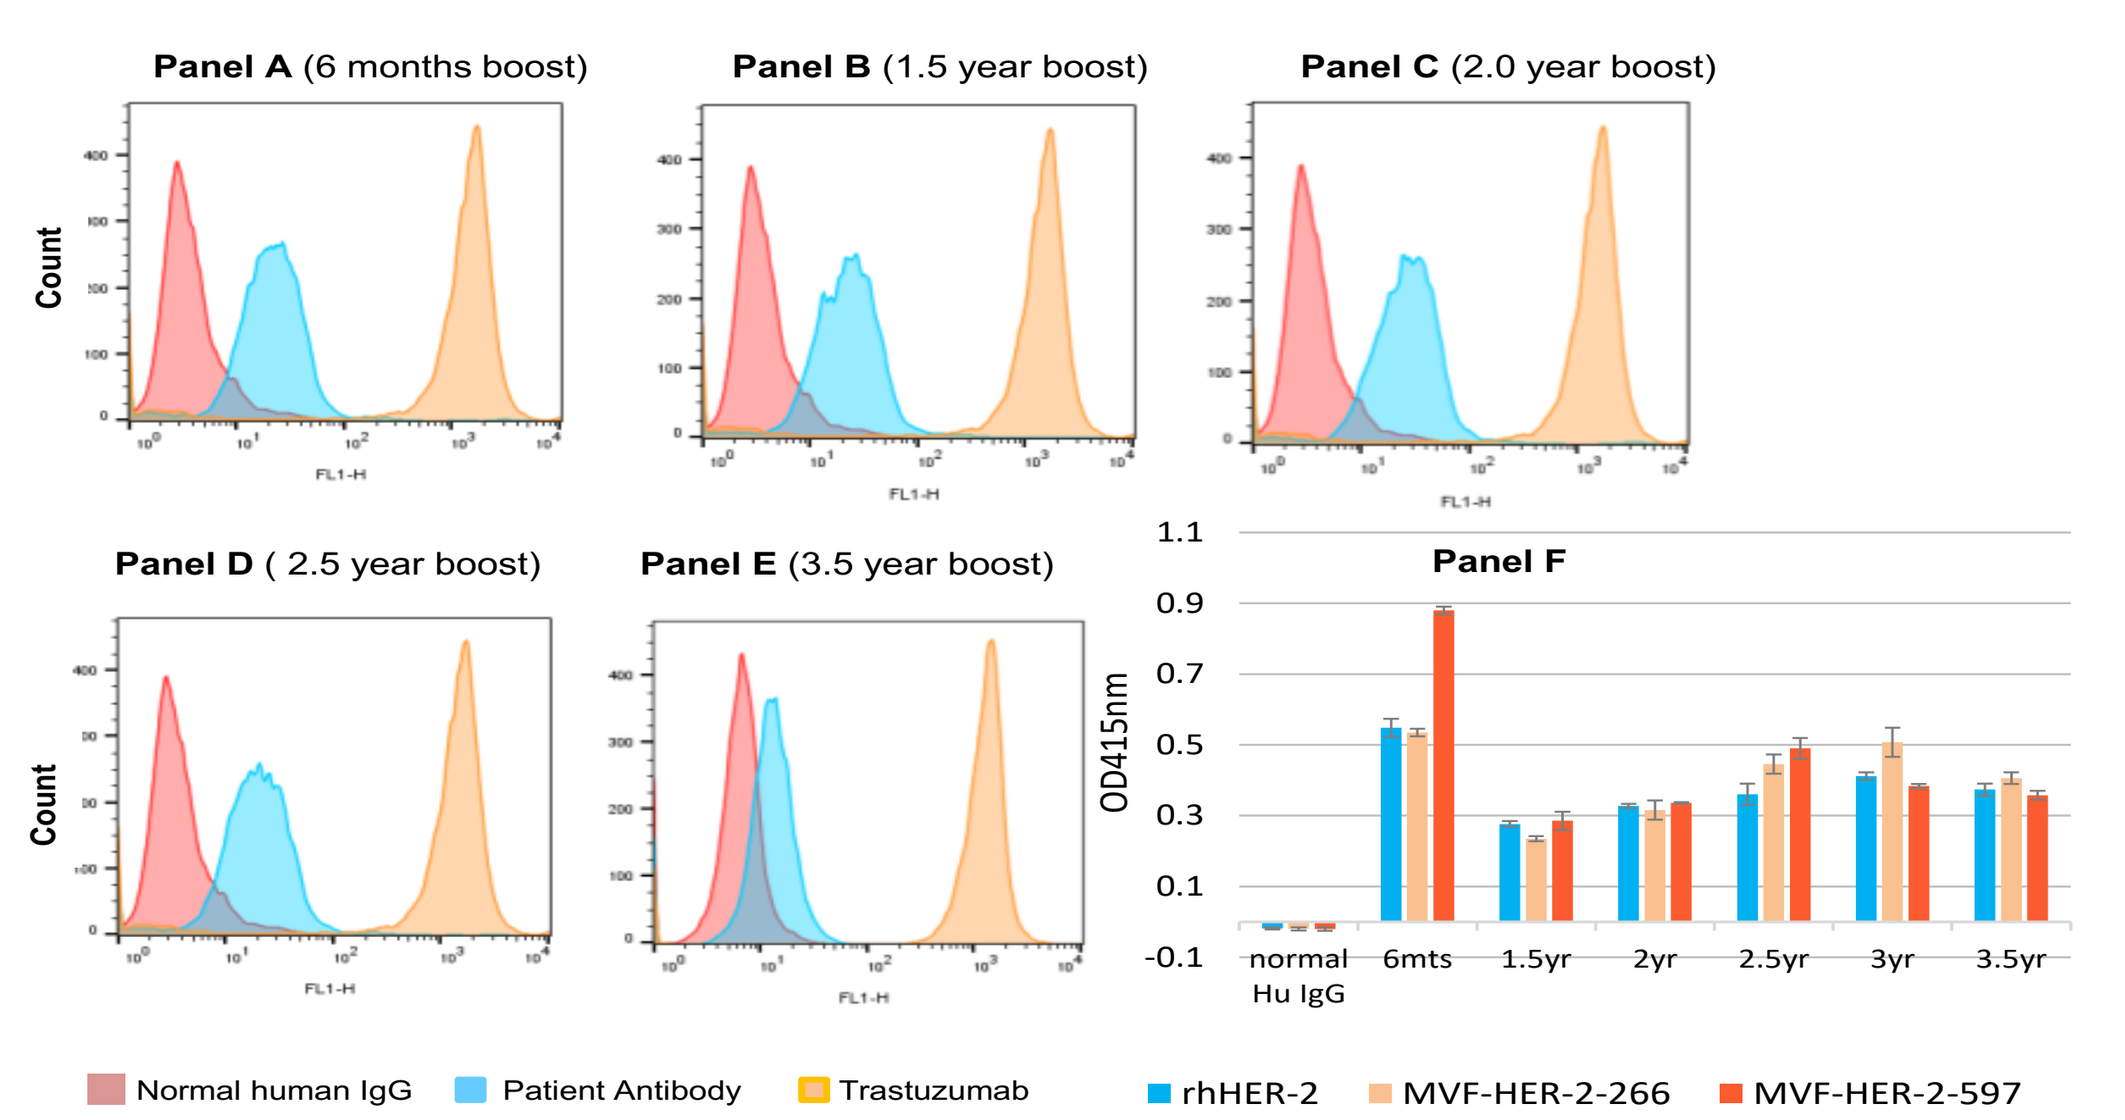
Panel A (6 months boost)
Panel B (1.5 year boost)
Panel C (2.0 year boost)
Panel D (2.5 year boost)
Panel E (3.5 year boost)
Panel F
Count
OD415nm
Normal human IgG
Patient Antibody
Trastuzumab
rhHER-2
MVF-HER-2-266
MVF-HER-2-597

Fig. 4 Panels (A-E) Flow cytometry of patient 2C antibodies by multiple boosts binding to BT-474. 5x10^5 BT-474 cells were incubated with 500µg/ml patient antibodies (blue), 500µg/ml control antibody (normal human IgG; red) and 50µg/ml commercial human anti-HER2 (Trastuzumab; yellow) in staining buffer (PBS+0.05%BSA+0.2% NaN_3) respectively at 4°C for 2 hours and then with 1.6 µg/ml Alexa Fluor® 488 anti-human IgG at 4°C for 45min. The stained cells were analyzed on a FACS Caliber machine. Panel F shows immunogenicity/antigenicity against vaccine and recombinant HER-2

Fig. 5 Effects of purified Abs on proliferation, phosphorylation, apoptosis, and ADCC

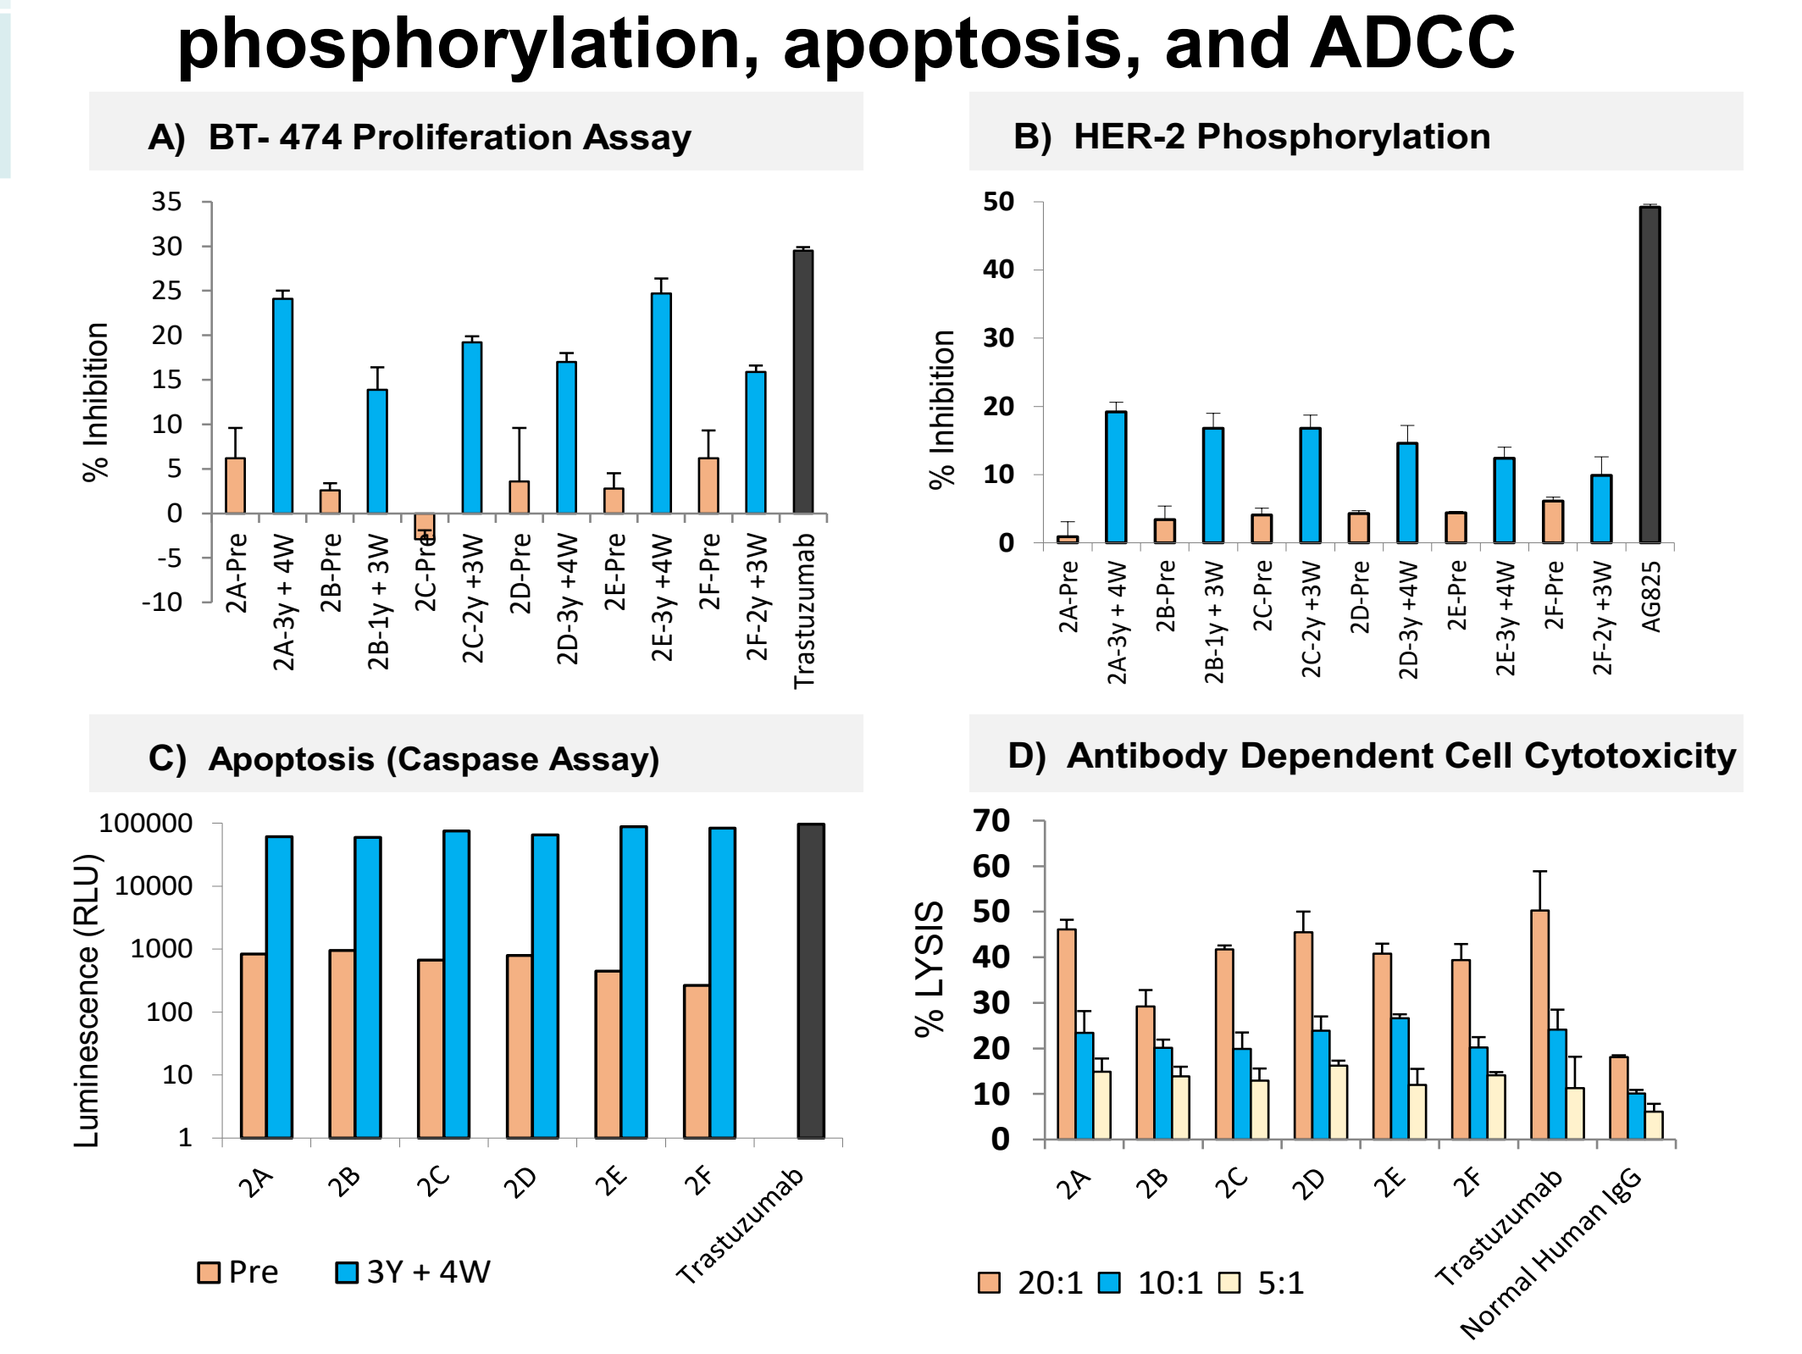
A) BT- 474 Proliferation Assay
B) HER-2 Phosphorylation
C) Apoptosis (Caspase Assay)
D) Antibody Dependent Cell Cytotoxicity
% Inhibition
Luminescence (RLU)
% LYSIS
Pre
3Y + 4W
20:1
10:1
5:1

Figure 5. Mechanistic assays: A) Proliferation assay was performed with BT-474 (10x 10^4 cells/ well). Inhibition % was calculated as previously described (see References). Trastuzumab served as positive control and preimmune sea served as negative control. **B) HER-2 phosphorylation** was performed by using BT474 (1x10^6 cells/ well) incubated with patient antibodies. Phosphorylation was determined by Duoset IC (R&D systems) for total human phospho-HER-2 according to manufacturer directions. **C) Apoptosis** was measured using the caspase activity assay (CaspaseGlo; Promega) BT-474 breast cancer cells were treated with the vaccine antibodies and caspase activity per manucfacturer's instructions. Data shows average of three different experiments following treatment with patient antibodies from dose level 2. **D) ADCC** was by using the bioluminescence non-radioactive cytotoxicity assay (aCella-Tox kit; Cell technology) according to manufacturer's recommendations. BT474 target cells were incubated with different amounts of effector cells (PBMC's) after treatment with patient antibodies. Trastuzumab was used as positive control, and normal human IgG was used as negative control.
Garrett JT, and Kaumaya et al., J Immunol 2007;178953,7120-31
AllenSD, and Kaumaya et al., J Immunol 2007;179:957,472-82

Conclusions

- Forty-nine patients with a median of 4 prior lines of chemotherapy received at least 1 vaccination. Twenty-eight patients completed the 3 vaccination regimen. Six patients received 1 six-month boost after the regimen, and one patient received 7 six-month boosts. No serious adverse reactions or dose-limiting toxicities were observed.
- The vaccine was well tolerated with dose level 2 defined as the OID/OBD and as the recommended phase II dose.
- The most common related toxicity in all patients was injection site reactions (24%).
- Two patients had a partial response (PR), 14 had stable disease (SD), and 19 had progressive disease (PD).
- The combination vaccine showed effectiveness in eliciting antibody responses without serious adverse reactions, treatment discontinuation due to unacceptable toxicities, DLTs, or evidence of treatment resistance.
- The study vaccine is safe, exhibits anti-tumor activity and shows preliminary indication that peptide vaccination may avoid therapeutic resistance and offer a promising alternative to monoclonal antibody therapies.
- Given the initial promise, continuous development of the vaccine is ongoing Phase 2 at the suggested OBD in a less heavily pretreated patient population in breast and/or gastrointestinal malignancies (gastroesophageal and colorectal) with HER-2/EGFR overexpression at the James Cancer Hospital, Columbus, Ohio
- **The Combination HER-2 vaccine has been licensed by IMUGENE for further development**

References

1. Bekaii-Saab T, and Kaumaya, PTP *et al*, Clin Cancer Res 2019 online Feb 25., DOI: 10.1158/1078-0432 (CCR-18-3997).
2. Kaumaya PT et al, Phase I active immunotherapy with combination of two chimeric, human epidermal growth factor receptor 2, B-cell epitopes fused to a promiscuous T-cell epitope in patients with metastatic and/or recurrent solid tumors. J Clin Onc 2009, 27:5270-7
3. Kaumaya PTP. Human Vaccines & Immunotherapeutics, 2015:11(6):1368-86

Acknowledgements

Funding NIH R01 CA CA84356 & NIH R21 CA13508 to PTPK